GlaxoSmithKline
Oncology

Poster #3140

Decrease in Serum Extracellular Domain of HER2 at 4 and 8 Weeks Is Associated With Prolonged Progression-Free Survival on Lapatinib Monotherapy

Allan Lipton[1], Kim Leitzel[1], Suhail Ali[2], Walter Carney[3], Greg Platek[4], Klaudia Steplewski[4], Ron Westlund[4], Robert Gagnon[4], Lisa O'Rouke[4], Anne-Marie Martin[4], Julie Maltzman[4]

[1]Medical Oncology/Internal Medicine, Milton S. Hershey Medical Center, Hershey, PA, United States, 17033-0850; [2]Lebanon Veterans Administration, Lebanon, PA, United States, 17042;
[3]Oncogene Science, Siemens Healthcare Diagnostics Inc., Cambridge, MA, United States, 02142; [4]Medicine Development Center, GlaxoSmithKline, Collegeville, PA, United States, 19426-0989.

INTRODUCTION

- HER2 receptor is overexpressed or amplified in ~25% of primary breast carcinomas and is associated with more aggressive disease and poor prognosis[1]
- The extracellular domain (ECD) of the HER2 is a p97-115kDa fragment which can be proteolytically cleaved from the cell membrane of both normal and cancerous cells and released to the circulation[2,3]
- Elevated serum HER2 ECD levels are reported in 30% to 90% of patients with metastatic breast cancer (MBC)[4,5]
- Rising serum HER2 ECD levels have been associated with progressive MBC and poor response to endocrine therapy and chemotherapy[6]
 - Elevated pretreatment serum HER2 ECD levels are associated with decreased response to both first- and second-line endocrine therapies[6]
- Studies have shown that significant decrease of ≥20% in serum HER2 ECD levels from pretreatment levels was a predictor of outcome to trastuzumab therapy including disease-free and overall survival[7,8]
- Serum HER2 ECD levels can be accurately and reproducibly measured with an FDA-approved test intended for follow-up and monitoring of MBC patients whose initial serum HER2 ECD levels were above 15 ng/mL[8]
- Lapatinib is an oral, selective, reversible, small molecule tyrosine kinase inhibitor of both HER1 and HER2, and in combination with capecitabine, is an approved regimen for patients with HER2-positive MBC who received prior treatment with anthracyclines, taxanes, and trastuzumab[9]
- Lapatinib monotherapy or in combination with chemotherapy or other targeted agents has shown good activity as treatment for HER2-positive MBC[9]
- This study evaluated serum HER2 ECD levels and their association with progression-free survival (PFS) in patients receiving lapatinib monotherapy as first-line treatment for HER2-positive MBC

METHODS

- Serum samples were collected at baseline and every 4 weeks for 16 weeks
- Serum HER2 ECD levels were quantified using a commercially available serum HER2/neu test (Siemens Healthcare Dx/Oncogene Science)
- Serum HER2 ECD levels ≥15 ng/mL were considered to be elevated
- A significant serum HER2 ECD decline was defined as a decrease of ≥20% from baseline at the follow-up visit and derived based on 2.5 times the CV of the HER-2/neu assay
- Efficacy was assessed at Weeks 8 and 12 using RECIST criteria
- Kaplan-Meier analysis and graphs were generated using SPSS (Statistical Package for the Social Sciences for Windows [version 11.0])
- Cox Proportional Hazards models were performed using SAS (version 9.2)

Figure 1. EGF20009 Trial Design

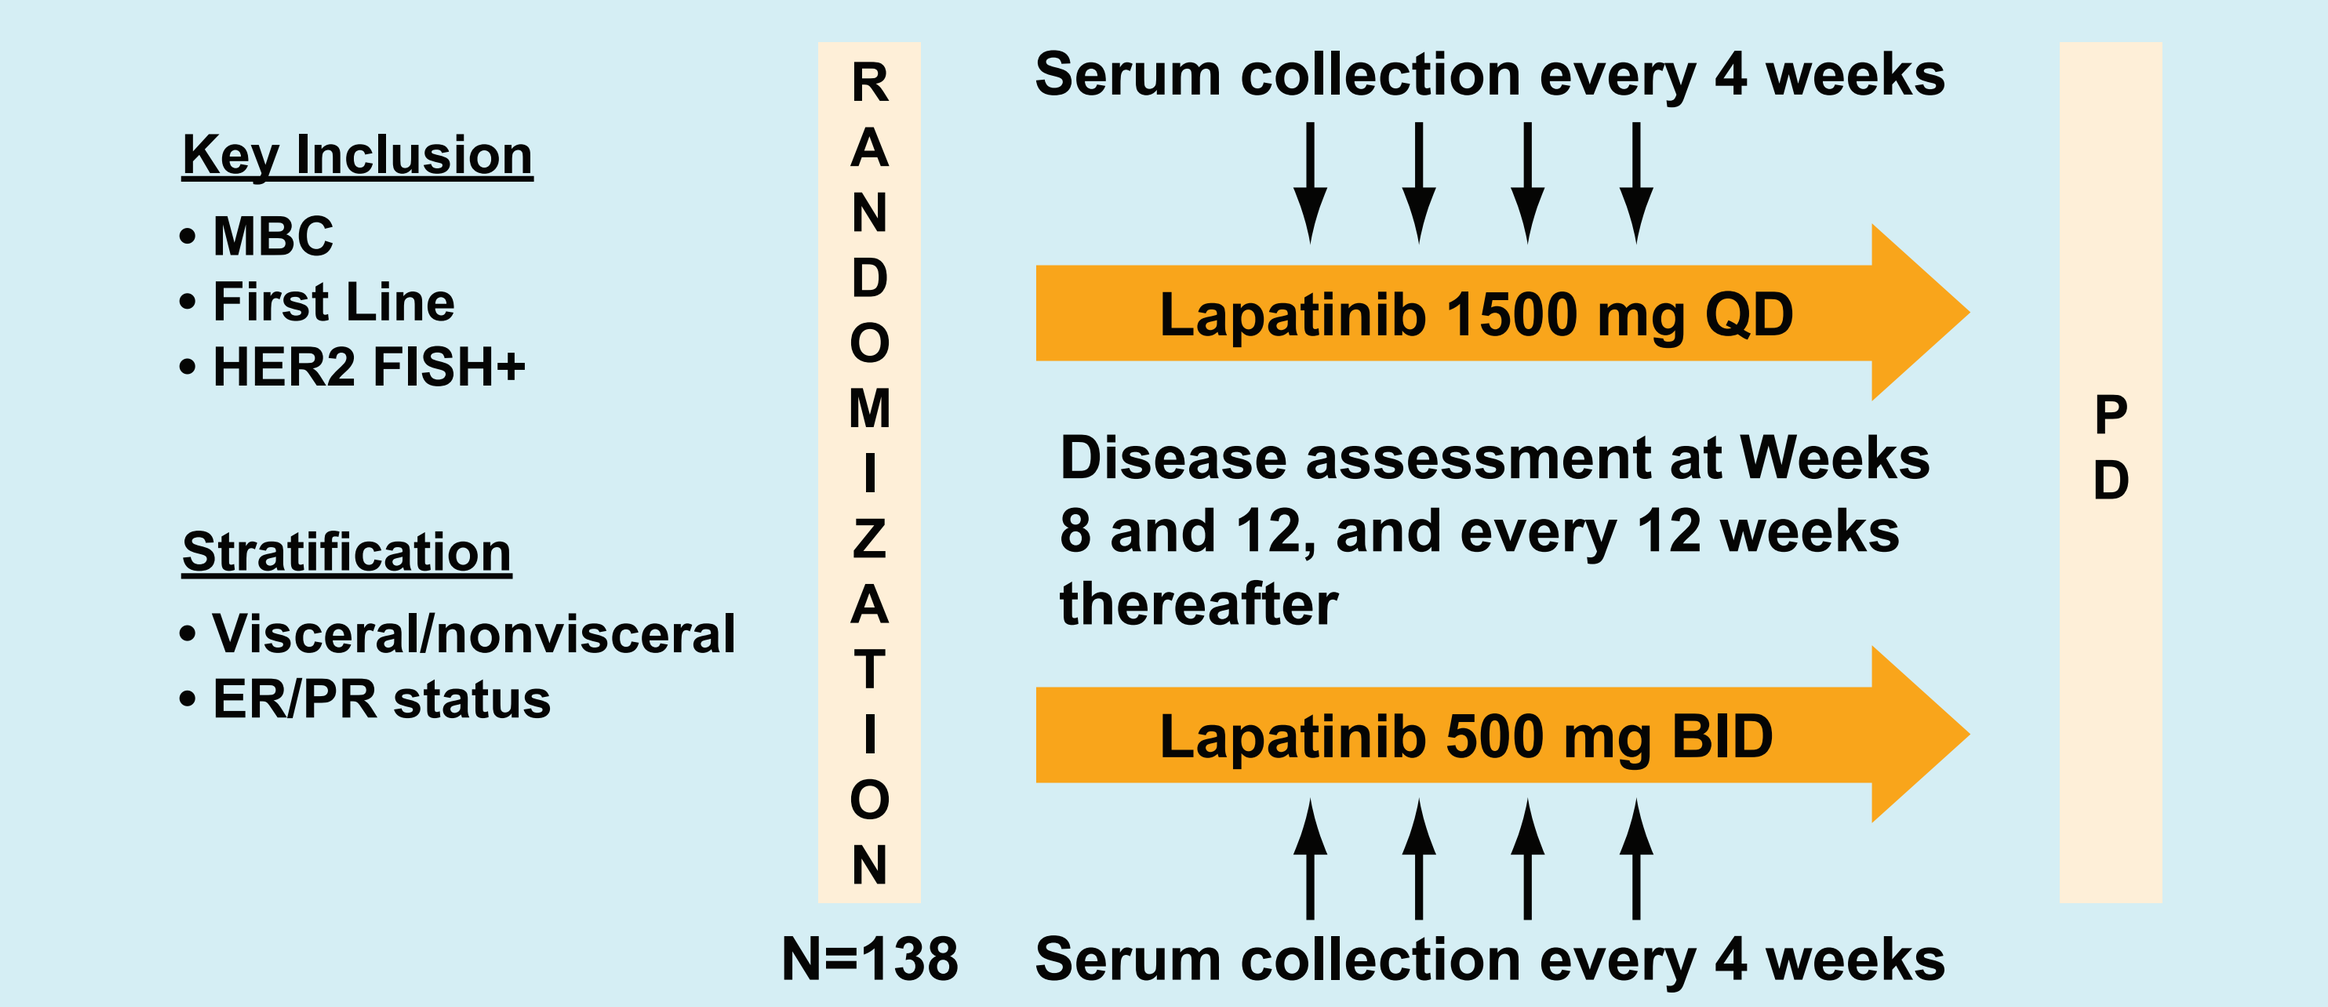
Key Inclusion
• MBC
• First Line
• HER2 FISH+
Stratification
• Visceral/nonvisceral
• ER/PR status
RANDOMIZATION
N=138
Serum collection every 4 weeks
Lapatinib 1500 mg QD
Disease assessment at Weeks 8 and 12, and every 12 weeks thereafter
Lapatinib 500 mg BID
Serum collection every 4 weeks
PD

FISH=fluorescence in situ hybridization; PD=progressive disease.

RESULTS

- 109/138 patients (79%) had elevated baseline serum HER2 ECD levels (above normal cutoff of 15 ng/mL), (data not shown); however, baseline serum HER2 was not associated with PFS
- 17/138 patients (12%) had no significant change in serum HER2 ECD levels (≥20% change from baseline; either a ≥20% increase or a ≥20% decrease)
- 64/138 patients (46%) had a serum HER2 ECD decrease of ≥20% by Week 12. None of the patients had a decrease in serum HER2 ECD after Week 12
- 64/138 (46%) patients had a serum HER2 ECD increase of ≥20% by Week 16
- Patients who achieved a ≥20% **decrease** in serum HER2 ECD at Week 4 or Week 8 had a significantly **longer** median PFS compared with patients who did not. Significantly longer PFS was also observed at any time during the first 16 weeks with median PFS of 246 days (95% CI 149, 254, N=64) for patients who achieved ≥20% **decrease** compared with 115 days (95% CI 82, 141, N=70) for patients who did not (*P*<.0001)
- Patients who achieved a ≥20% **increase** in serum HER2 ECD at any time during the first 16 weeks had a significantly **shorter** median PFS of 115 days (95% CI 100, 141, N=64) compared with 199 days (95% CI 145, 254, N=70) for patients who did not (*P*<.0001)
- Data from the Cox Proportional Hazards regression analysis suggests that minimum percentage change in serum HER2 ECD levels is an independent predictor of PFS after adjusting for minimum percentage change in tumor burden; however, further confirmation of these results in a larger trial is required

Table 1. Serum HER2 ECD Change in Patients Treated With Lapatinib Monotherapy

(First occurrence of ≥20% decrease or ≥20% increase from baseline level of serum HER2 ECD)

	Week 4	Week 8	Week 12	Week 16	Total*
Patients with decrease†, n	53	8	3	0	64
Patients with increase‡, n	42	16	3	3	64

*Total includes patients who did not maintain the ≥20% decrease or increase in HER2 ECD or data was missing after the initial occurrence of HER2 ECD change.
†74 patients did not achieve ≥20% decrease of HER2 ECD levels during the study.
‡74 patients did not achieve ≥20% increase of HER2 ECD levels during the study.

Figure 2. Progression-Free Survival for Patients Who Achieved at Least 20% Decrease in Serum HER2 ECD From Baseline at Week 4

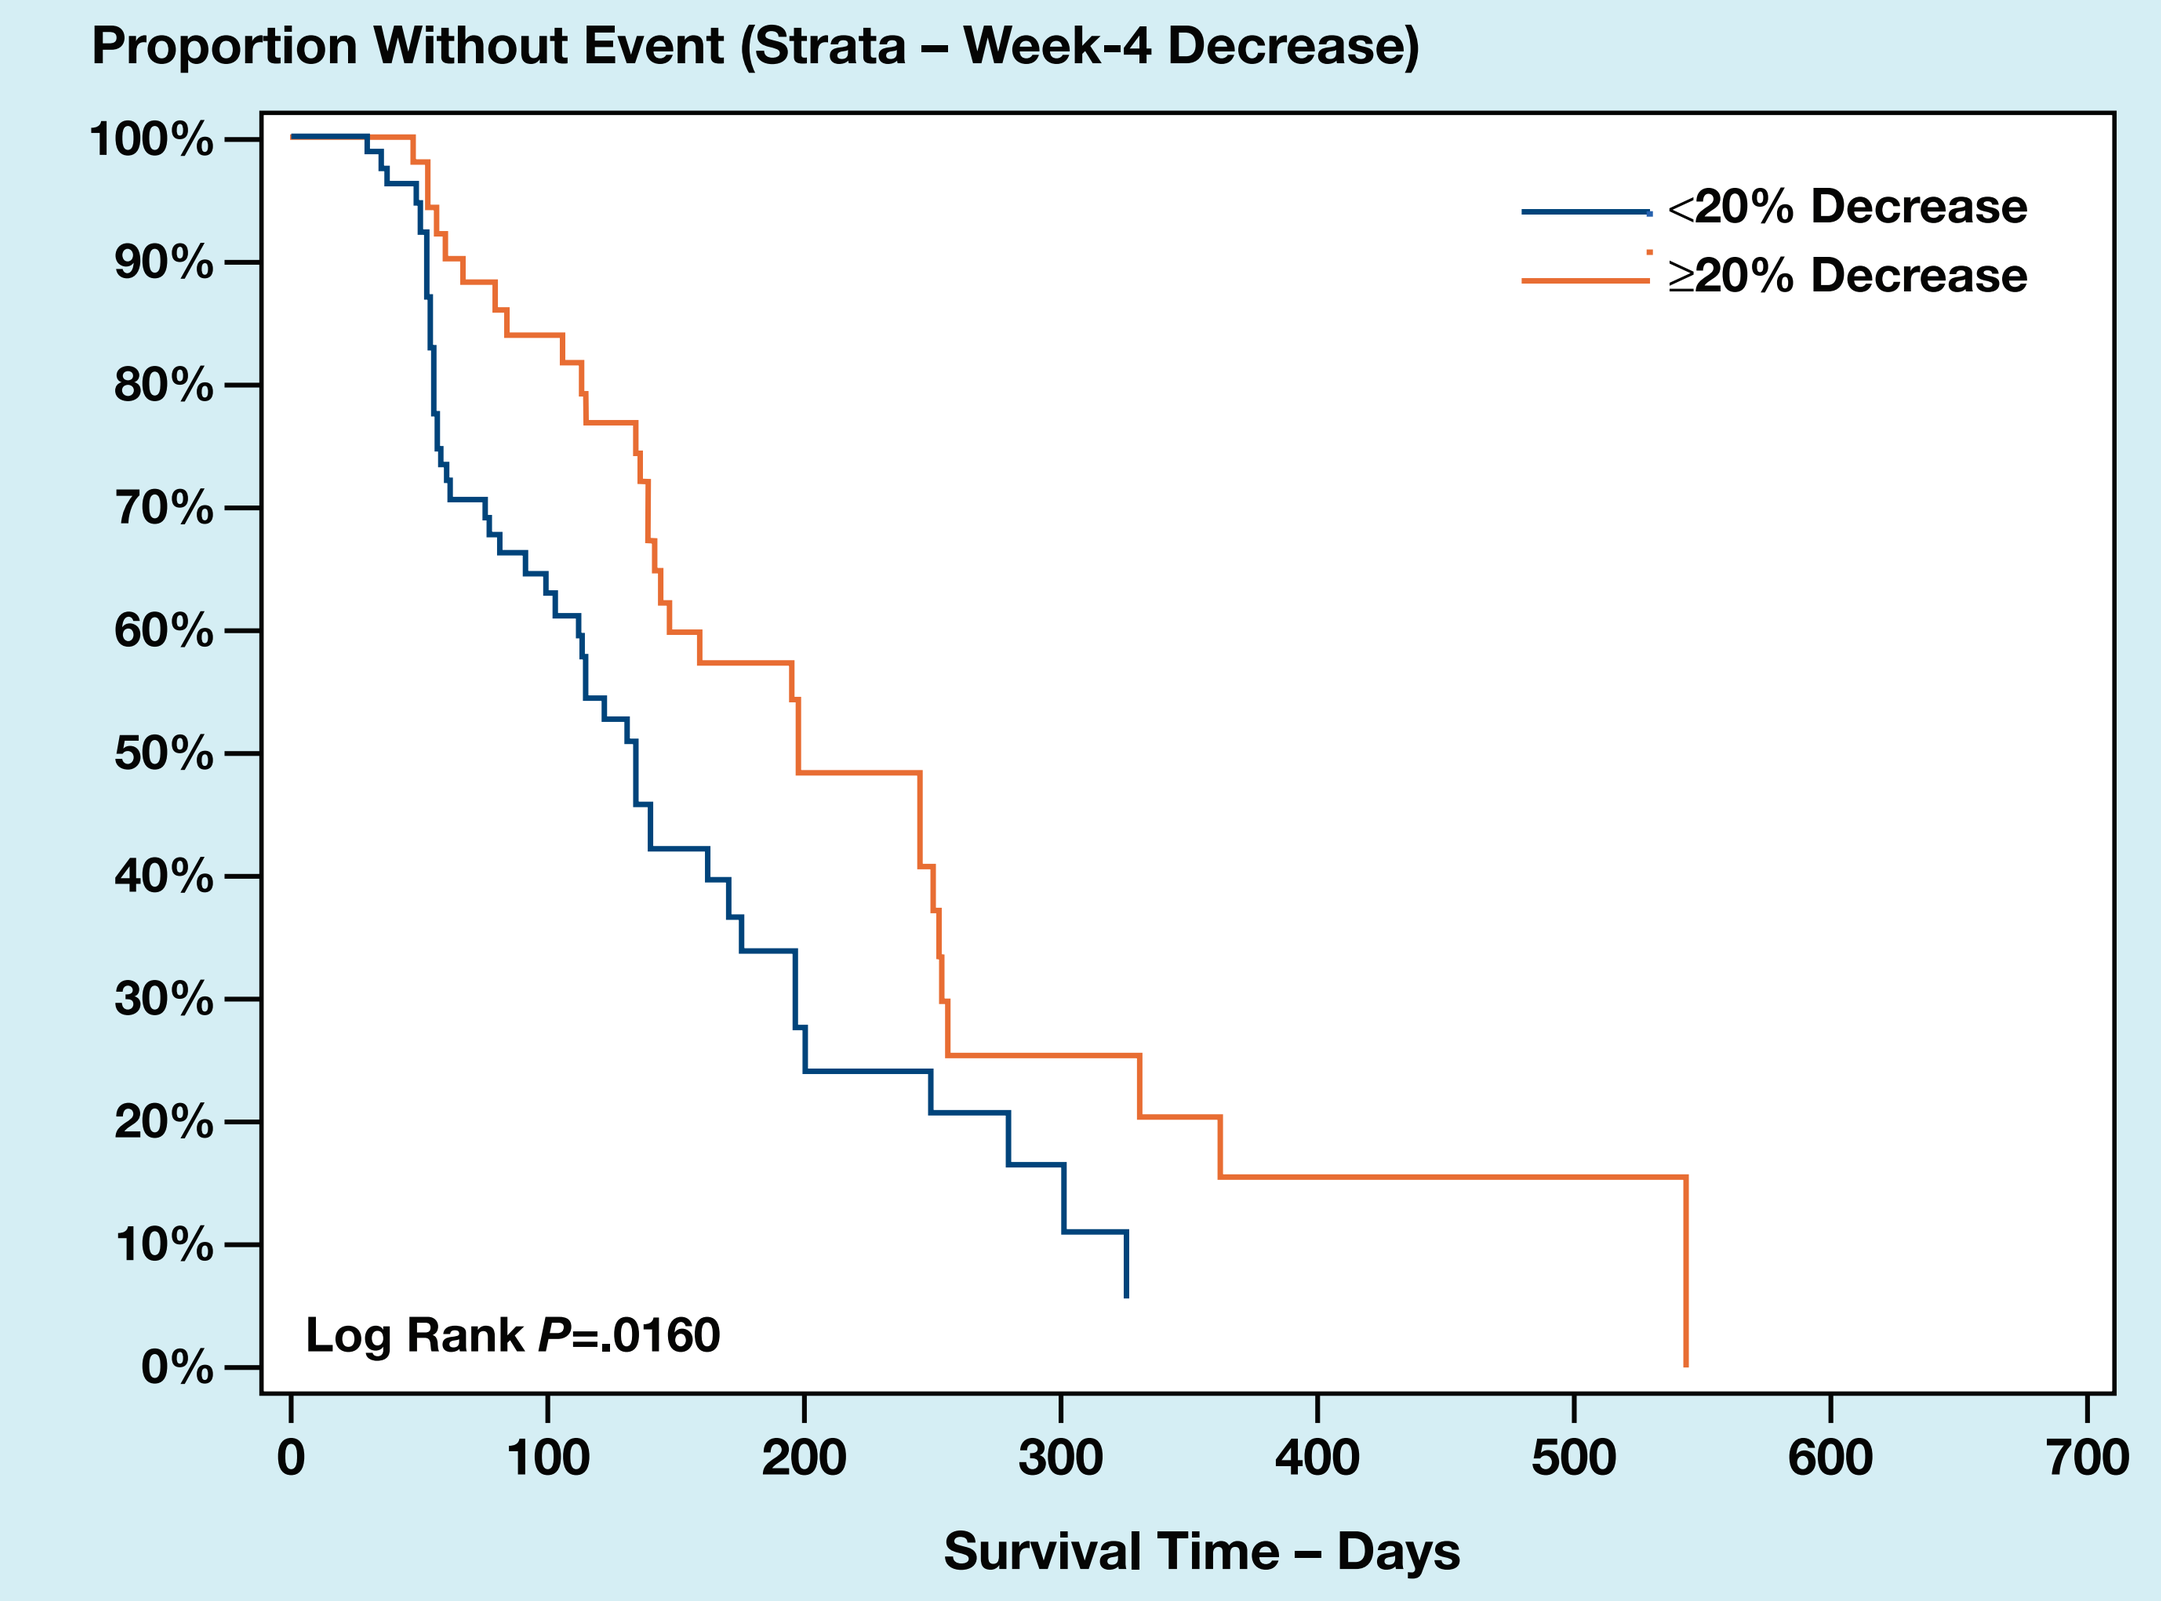
Proportion Without Event (Strata – Week-4 Decrease)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
<20% Decrease
≥20% Decrease
Log Rank P=.0160
0
100
200
300
400
500
600
700
Survival Time – Days

Week-4 Decrease	No. Subjects	Event	Censored	Median PFS	95% Confidence Interval
<20% Decrease	78	63% (49)	37% (29)	135	(104, 171)
≥20% Decrease	53	58% (31)	42% (22)	199	(145, 253)

Figure 3. Progression-Free Survival for Patients Who Achieved at Least 20% Decrease in Serum HER2 ECD From Baseline at Week 8

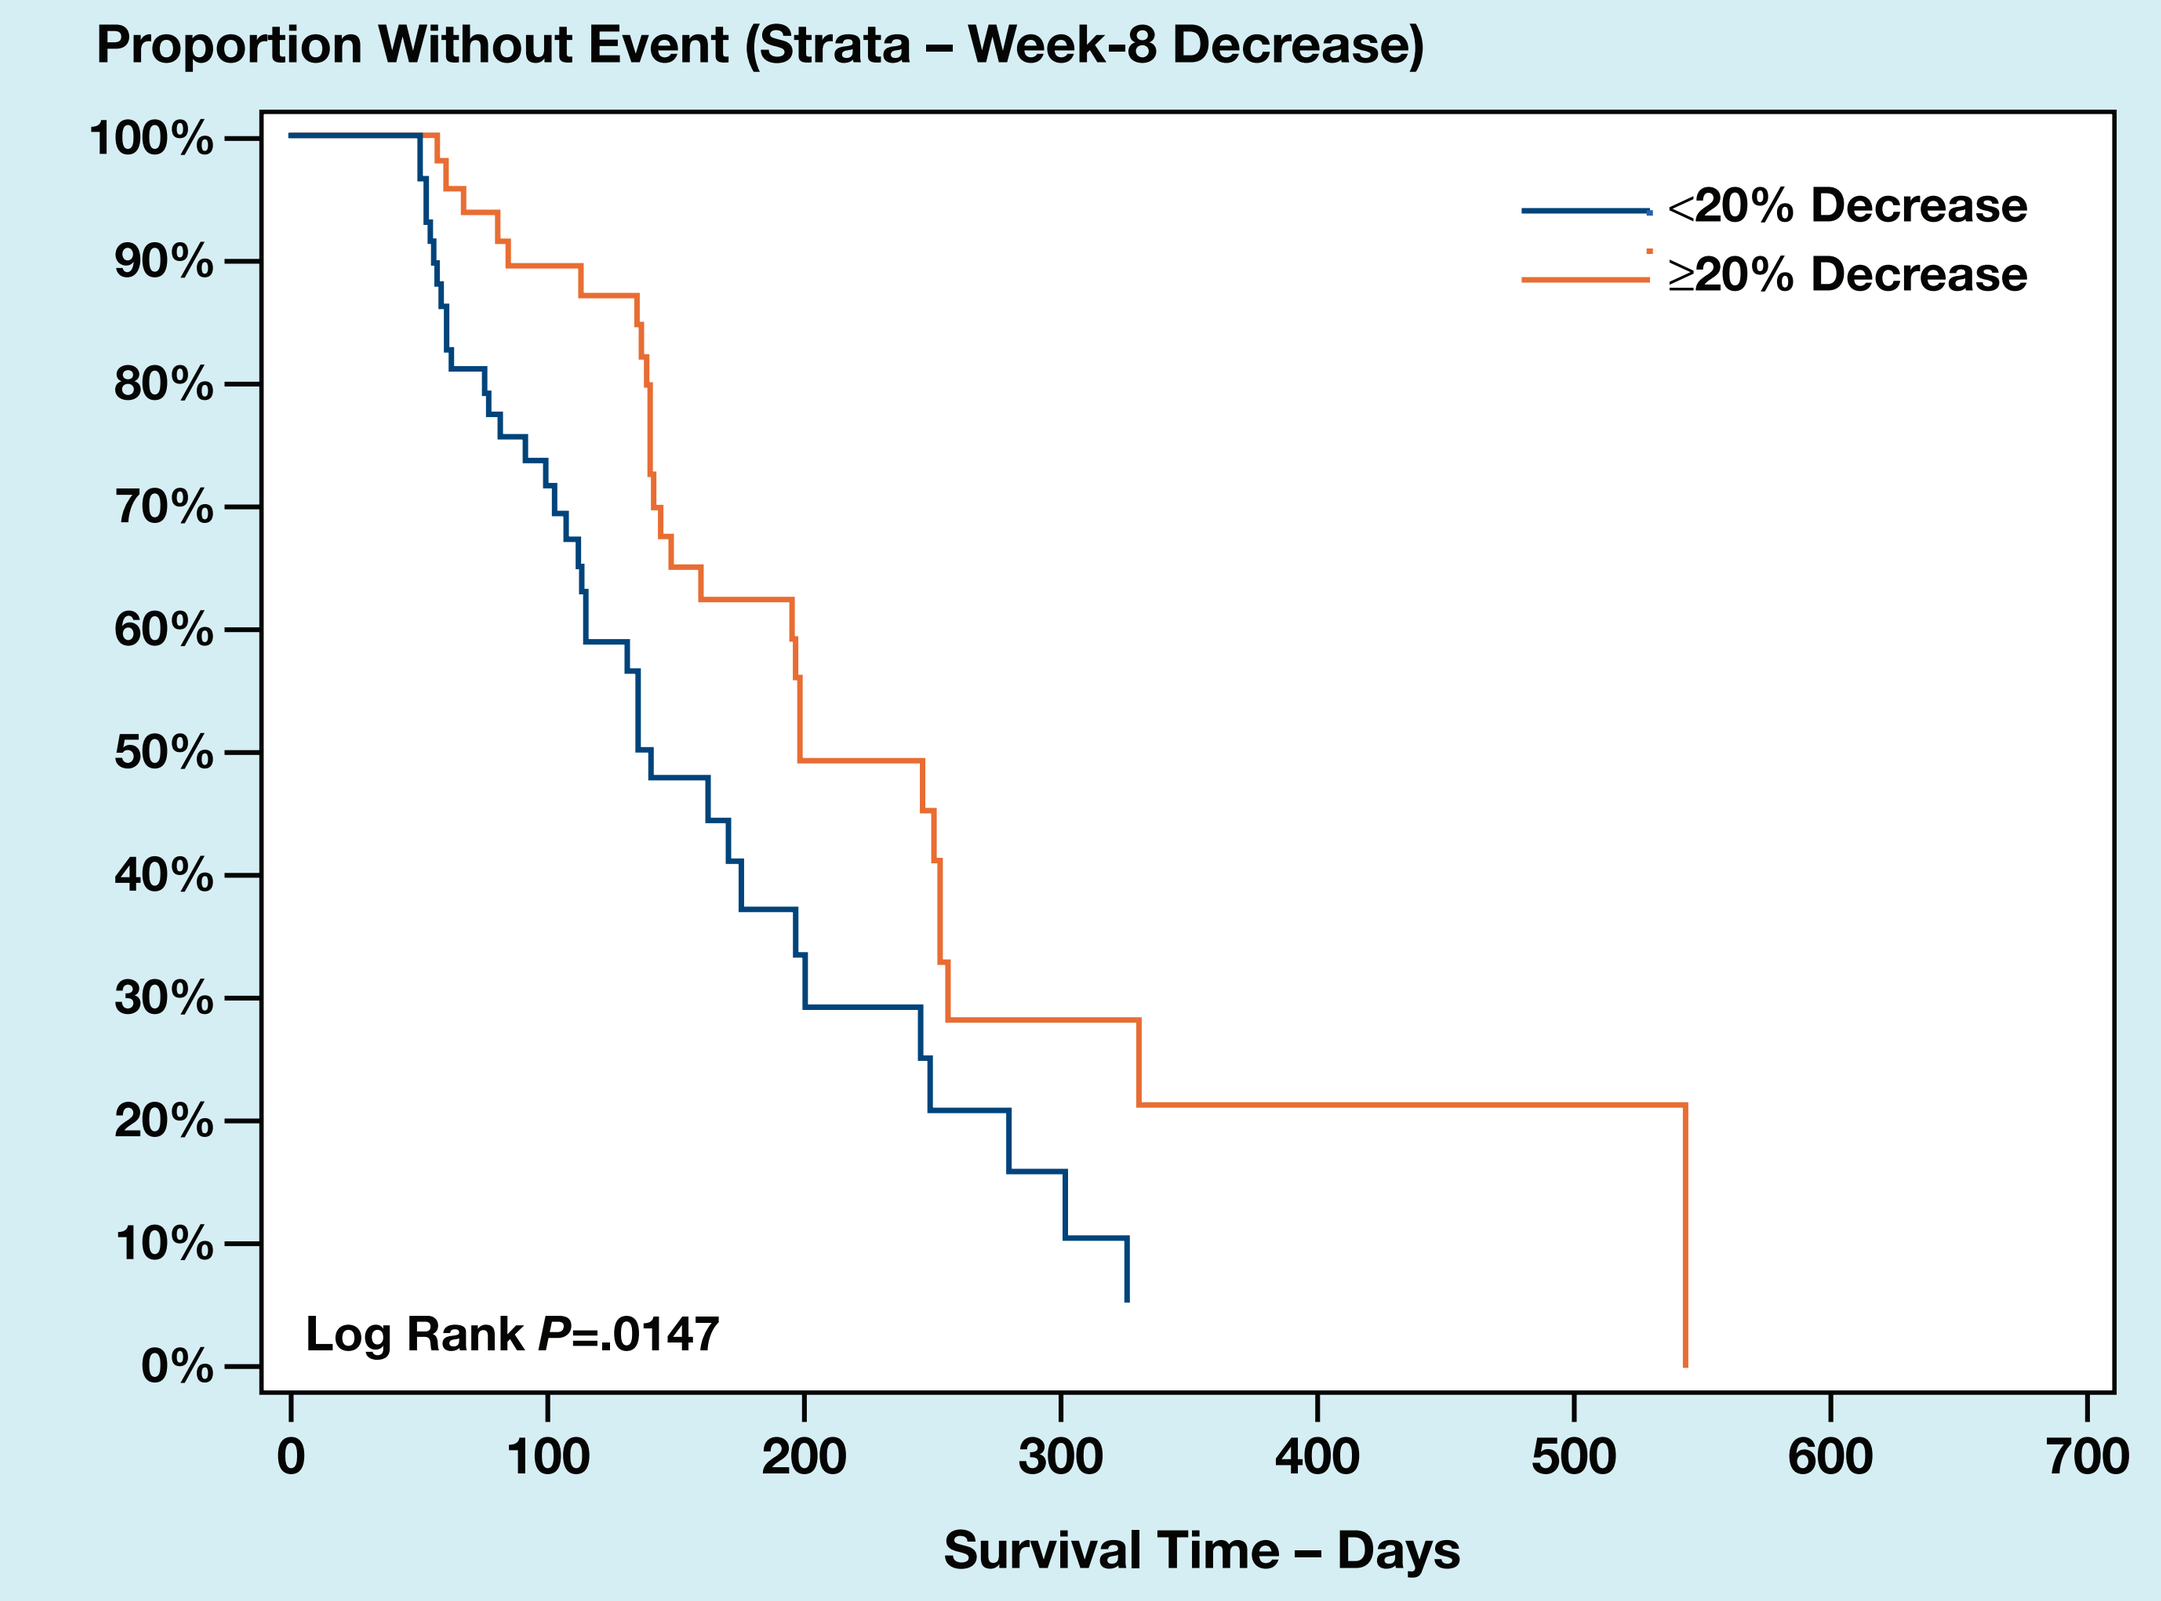
Proportion Without Event (Strata – Week-8 Decrease)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
<20% Decrease
≥20% Decrease
Log Rank P=.0147
0
100
200
300
400
500
600
700
Survival Time – Days

Week-8 Decrease	No. Subjects	Event	Censored	Median PFS	95% Confidence Interval
<20% Decrease	59	63% (37)	37% (22)	141	(115, 197)
≥20% Decrease	50	54% (27)	46% (23)	199	(160, 254)

Figure 4. Progression-Free Survival for Patients Who Achieved at Least 20% Decrease in Serum HER2 ECD From Baseline at Any Time by Week 16

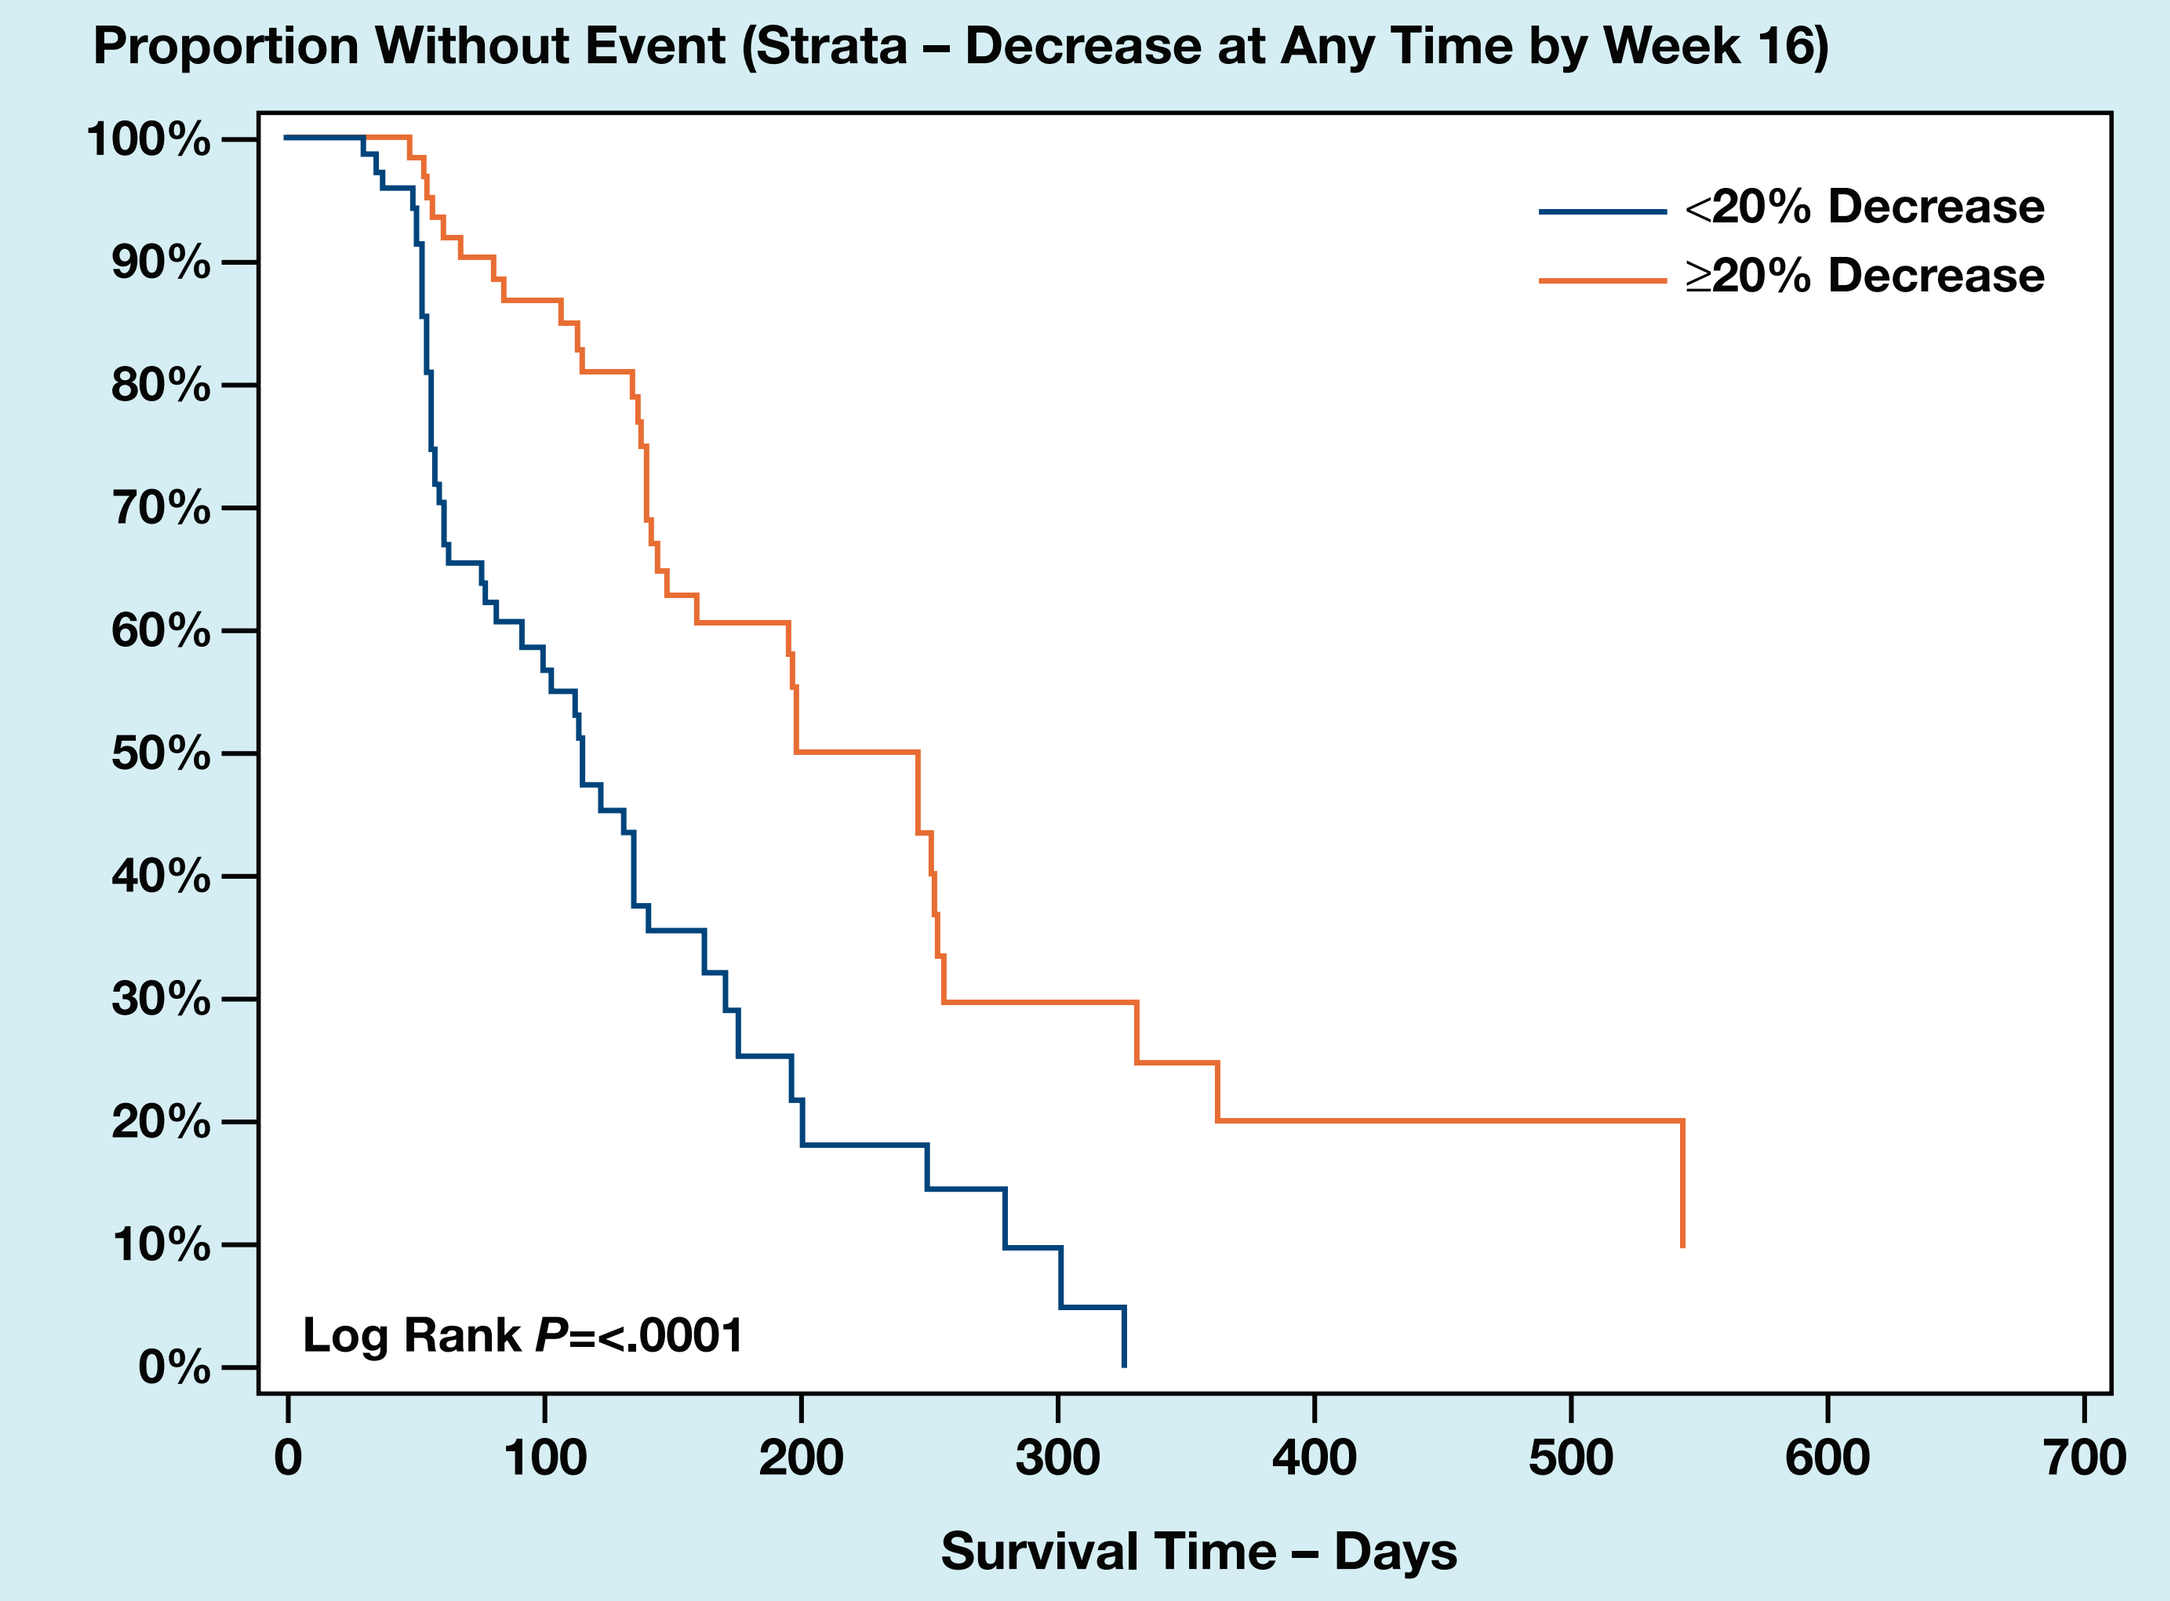
Proportion Without Event (Strata – Decrease at Any Time by Week 16)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
<20% Decrease
≥20% Decrease
Log Rank P=<.0001
0
100
200
300
400
500
600
700
Survival Time – Days

Anytime Decrease	No. Subjects	Event	Censored	Median PFS	95% Confidence Interval
<20% Decrease	70	69% (48)	31% (22)	115	(82, 141)
≥20% Decrease	64	53% (34)	47% (30)	246	(149, 254)

Figure 5. Progression-Free Survival for Patients Who Achieved at Least 20% Increase in Serum HER2 ECD From Baseline at Any Time by Week 16

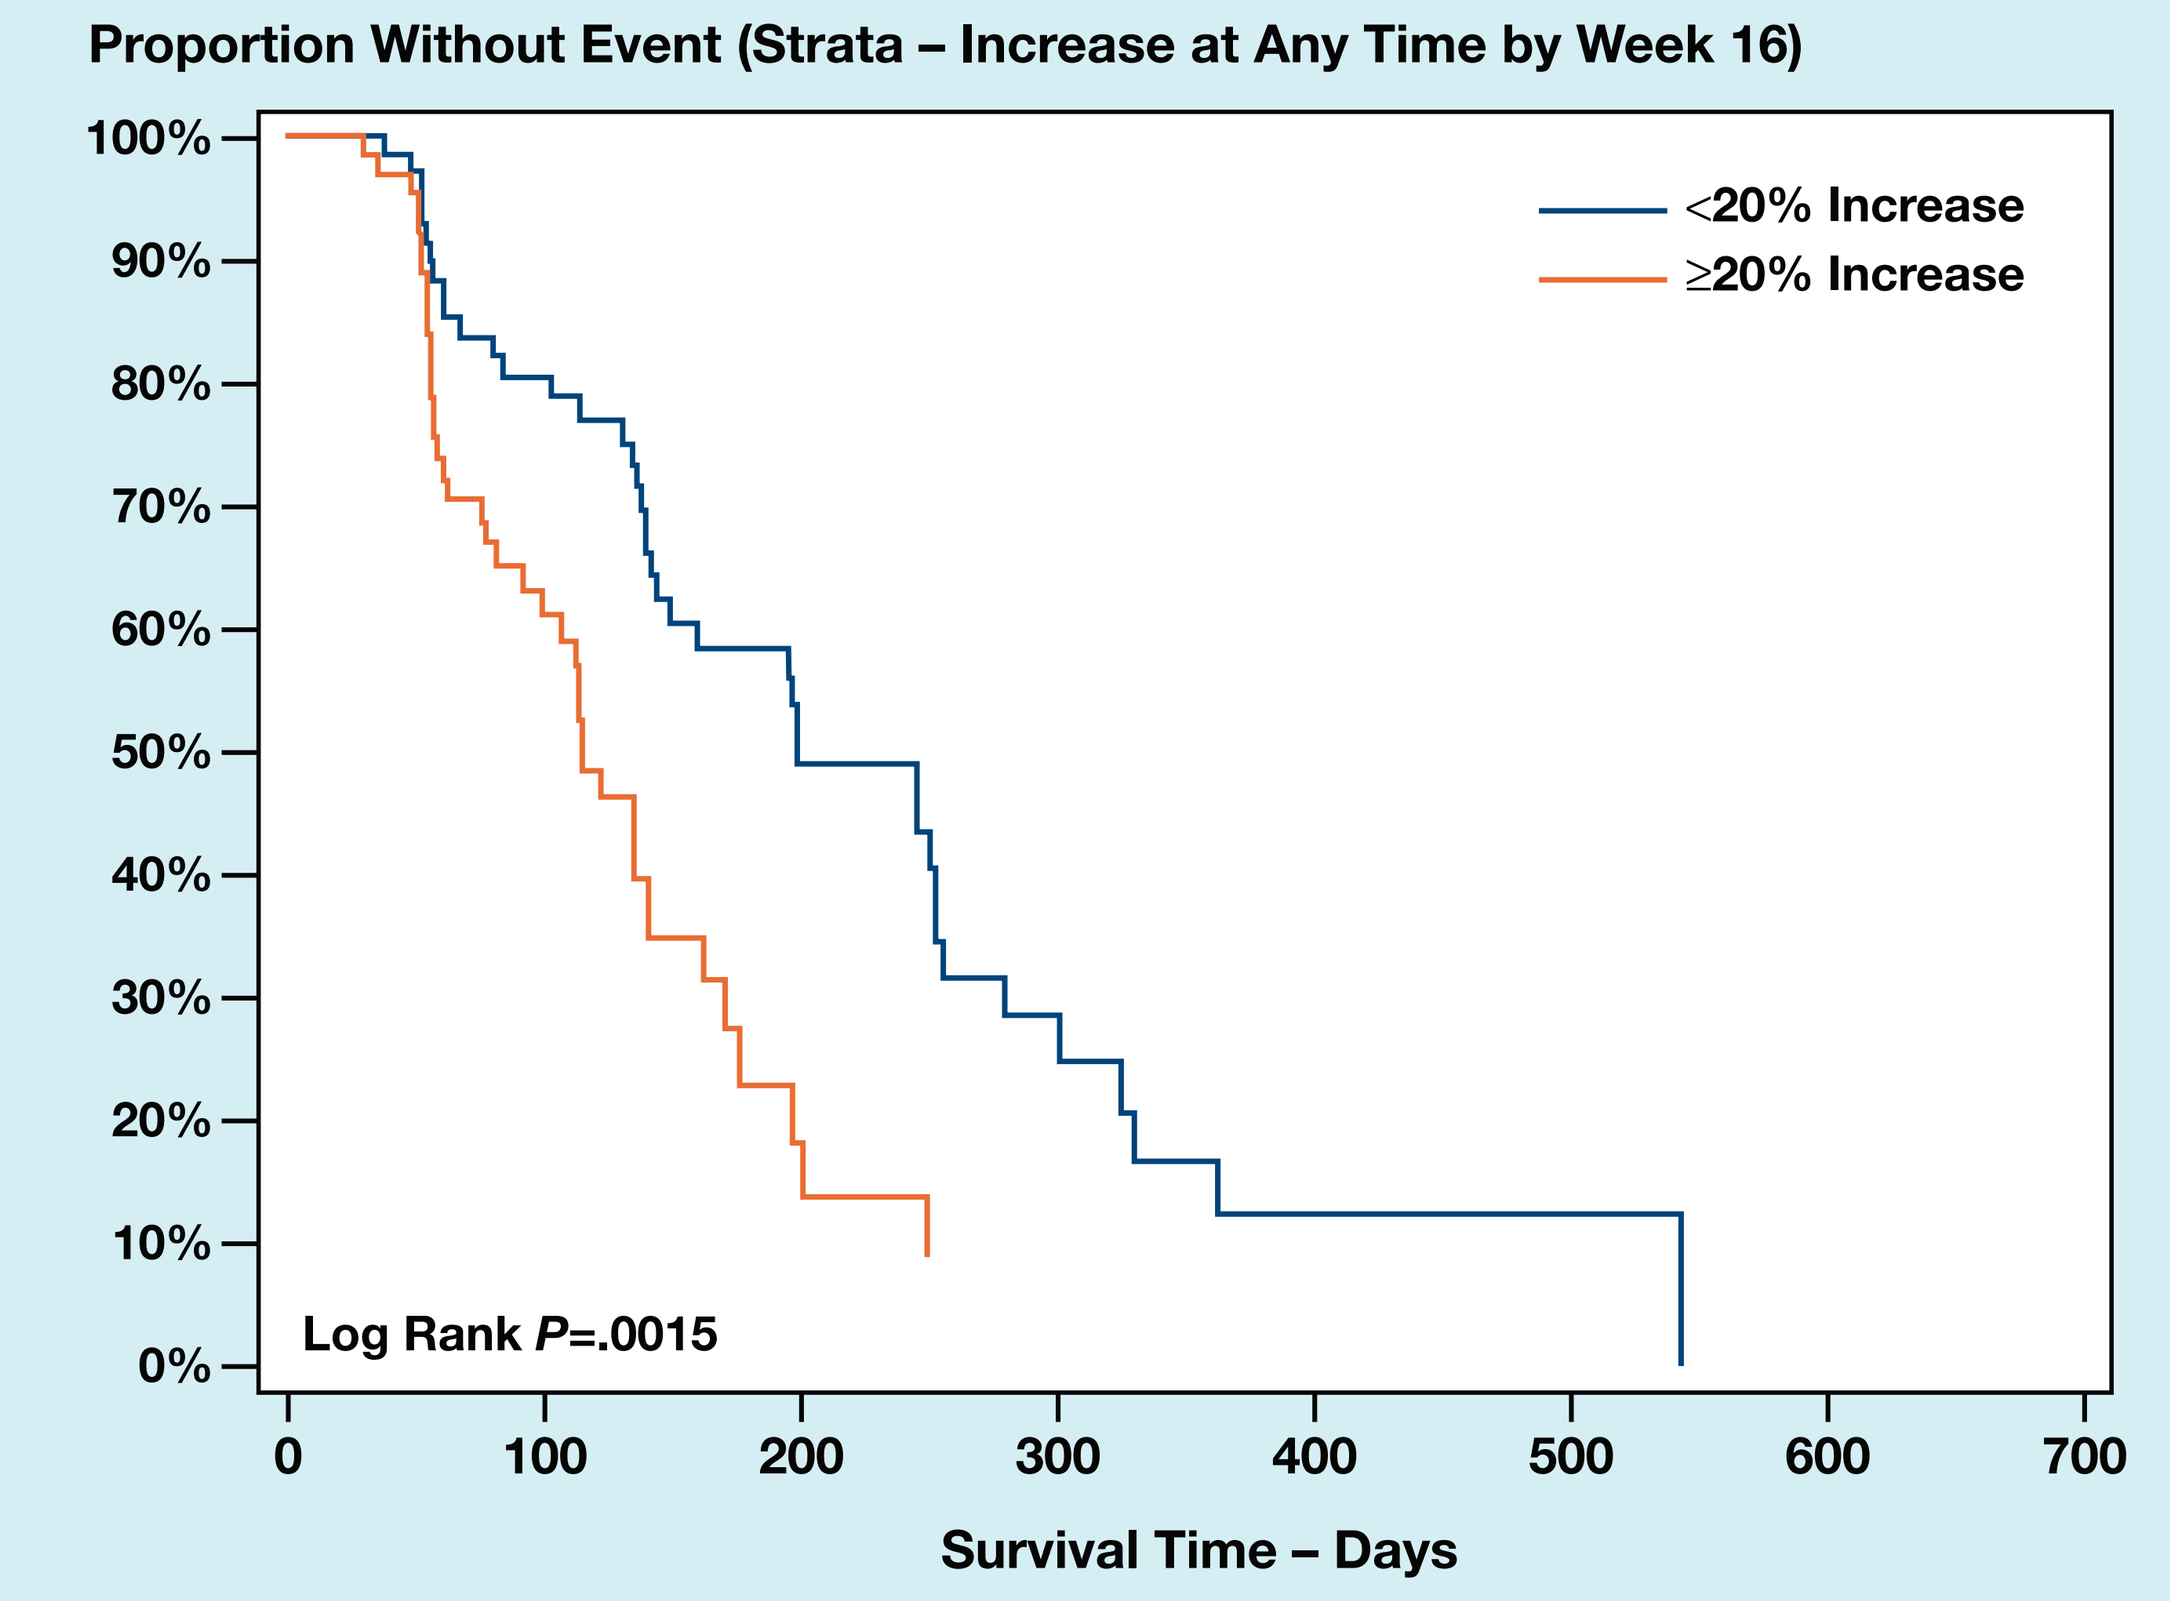
Proportion Without Event (Strata – Increase at Any Time by Week 16)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
<20% Increase
≥20% Increase
Log Rank P=.0015
0
100
200
300
400
500
600
700
Survival Time – Days

Anytime Increase	No. Subjects	Event	Censored	Median PFS	95% Confidence Interval
<20% Increase	70	59% (41)	41% (29)	199	(145, 254)
≥20% Increase	64	64% (41)	36% (23)	115	(100, 141)

Table 2. Cox Proportional Hazards with PFS

Multivariate Model Including Baseline Serum HER2 ECD, Baseline Tumor Lesion Size Measured by Sum of Longest Diameters (SLD), Minimum % Change From Baseline Serum HER2 ECD and Minimum % Change in Tumor Lesion SLD

Variable	Hazard Ratio	*P* value	Hazard Ratio 95% Confidence Limits	
Baseline ECD	1.001	0.1070	1.000	1.003
Baseline SLD	1.004	0.1244	0.999	1.009
Minimum* Percent Change ECD (first 16 weeks)	1.005	**0.0176**	1.001	1.010
Minimum† Percent Change SLD (first 16 weeks)	1.021	**0.0003**	1.010	1.033

*Minimum percent change ECD is the largest decrease or smallest increase (in the absence of any decreases) in serum HER2 ECD levels from baseline at any timepoint.
†Minimum percent change SLD is the largest decrease or smallest increase (in the absence of any decreases) of SLD from baseline at any timepoint.
Note: the timings of the ECD and SLD minimum percent changes may not be synchronized and therefore may warrant further investigation.

CONCLUSIONS

- At least 20% decrease from baseline of serum HER2 ECD levels is associated with longer PFS while on lapatinib monotherapy
- Further evaluation in larger clinical trial is required to determine, if changes in serum HER2 ECD levels may be used to monitor patients with HER2-positive MBC receiving lapatinib therapy as this study did not have a non-lapatinib containing treatment arm for proper comparison
- In addition, further evaluation is warranted to assess if changes in serum HER2 ECD levels can be used to not only monitor patients but potentially allow for selection of more individualized therapy

REFERENCES

1. Moy B, Goss PE. Lapatinib: current status and future directions in breast cancer. *Oncologist*. 2006;11:1047-1057.
2. Carney WP, Neumann R, Lipton A, Leitzel K, Ali S, Price CP. Potential clinical utility of serum HER-2/neu oncoprotein concentrations in patients with breast cancer. *Clin Chem*. 2003;49:1579-1598.
3. Carney WP, Leitzel K, Ali S, Neumann R, Lipton A. HER-2/neu diagnostics in breast cancer. *Breast Cancer Res*. 2007;9:207.
4. Hayes DF, Yamauchi H, Broadwater G, et al. Circulating HER-2/erbB-2/c-neu (HER-2) extracellular domain as a prognostic factor in patients with metastatic breast cancer: Cancer and Leukemia Group B Study 8662. *Clin Cancer Res*. 2001;7:2703-2711.
5. El-Sawy WH, Abou Taleb FM, Abdel Kader M, Dorgham YT, Radwan DA, Gharib AF. Circulation HER2 extracellular domain and response to chemotherapy in metastatic breast cancer. *J Egyptian Nat Cancer Inst*. 2002;14:29-37.
6. Bramwell VH, Doig GS, Tuck AB, et al. Changes over time of extracellular domain of HER2 (ECD/HER2) serum levels have prognostic value in metastatic breast cancer [Epub ahead of print]. *Breast Cancer Res Treat*. 2008.
7. Ali SM, Esteva FJ, Fornier M, et al. Serum HER-2/neu change predicts clinical outcome to trastuzumab-based therapy. *J Clin Oncol* 2006;24(suppl):500.
8. Esteva FJ, Cheli CD, Fritsche H, et al. Clinical utility of serum HER2/neu in monitoring and prediction of progression-free survival in metastatic breast cancer patients treated with trastuzumab-based therapies. *Breast Cancer Res*. 2005;7:R436-R443.
9. Cameron DA, Stein S. Drug Insight: intracellular inhibitors of HER2—clinical development of lapatinib in breast cancer. *Nat Clin Pract Oncol*. 2008;5:512-520.

This research was conducted by GlaxoSmithKline, Philadelphia, Pennsylvania, USA.
Tykerb and Tyverb are trademarks of the GlaxoSmithKline group of companies.